A dose selection rationale based on hemodynamics for sildenafil in pediatric patients with pulmonary arterial hypertension (PAH)

Pascal Chanu(1), Xiang Gao(2), Mike Smith(3), René Bruno(1), Lutz Harnisch(3)

(1) Pharsight, a CertaraTM company, St. Louis, MO, USA; (2) Pfizer, Clinical Pharmacology, New London, CT, USA; (3) Pfizer, Pharmacometrics, Sandwich, UK

BACKGROUND

The efficacy of treatments for PAH in adults is mainly based on improvements in exercise capacity i.e. the six-minute walk distance (6MWD) test.

In children ≥7 y, 6MWD is not well reproducible and peak oxygen consumption (pVO2) is used instead. In younger patients an exercise capacity test is not doable and hemodynamic endpoints may be used to assess efficacy [1, 2].

A recent FDA analysis [3] showed a relationship between changes from baseline in 6MWD and an hemodynamic endpoint i.e. pulmonary vascular resistance index (PVRI) in the adult PAH population and that this relationships is consistent across drug classes. From this relationship, a 30% ΔΔ decrease (placebo corrected change from baseline) in PVRI corresponds to a 10-15% ΔΔ enhancement in 6MWD.

Sildenafil (REVATIO®), 20 mg TID, received approval for the treatment of adult PAH in the US based on 6MWD data.

Consistency of Sildenafil data for both adults and pediatrics with predictions from the FDA model has been assessed; Dose selection for sildenafil in adults is consistent across both hemodynamic and exercise based criteria [4].

OBJECTIVE

The objective of this analysis was to support the dose selection of sildenafil in the pediatric PAH patients using a model-based approach to pulmonary vascular resistance (PVR) outcomes, bridging efficacy from adults to children.

METHODS

A population PK/PD analysis of PVR data from two pivotal sildenafil trials in adult [5] (n=218) and pediatric patients [6] (n=219, 1-17 y) was performed.

A model was developed in NONMEM 7 to characterize the relationship between PVR, baseline patho-physiological covariates i.e. age, body surface area (BSA), PAH functional class, PAH etiology, ability for exercise capacity assessment and sildenafil average concentration at steady state (individual empirical Bayesian estimates) obtained using a previously developed population PK model [7]. The model was developed using an exploratory graphical analysis which suggested piece-wise linear relationships between covariates and baseline PVR, between exposure and drug effect, followed by a stepwise inclusion of covariates.

Simulations based on clinically defined success criteria to achieve similar hemodynamic responses in children as seen previously in adults under the labeled dose were conducted to support the dose selection in pediatric patients.

RESULTS

PVR was modeled as a function of baseline covariates (functional class, etiology, age, BSA, ability for exercise capacity assessment) and sildenafil exposure.

PVR at baseline

$$PRV_{i,base} = exp(Baseline)$$

$$Baseline_{adults} = BASE \cdot (1 + (BASE_{age} \cdot (AGE - 18) + BASE_{BSA} \cdot (BSA - 1.5) + BASE_{FC} \cdot FCL))$$

$$Baseline_{pediatrics} = BASE \cdot (1 + (BASE_{BSA} \cdot (BSA - 1.5) + BASE_{FC} \cdot FCL))$$

FCL taking the values -1,0,1 for subjects of functional class (FC) 1, 2, or 3 and 4, respectively

For adult patients having CTD (Connective Tissue Disease) as their etiology, BASE substituted with $BASE_{CTD}$

PVR at end of treatment

For $C_{av,ss} \le$ Thrs (threshold concentration) $PRV_{i,end} = exp(Baseline + E_0 + Slope_1 \cdot C_{av,ss})$

For $C_{av,ss} >$ Thrs $PRV_{i,end} = exp(Baseline + E_0 + Slope_1 \cdot Thrs + Slope_2 \cdot (C_{av,ss} - Thrs))$

E_0 placebo effect (disease worsening)

For developmentally able children, not having a shunt as their etiology, $Slope_1$ substituted with $Slope_{1,devable}$

$C_{av,ss}$ average concentration at steady state derived from the population PK model using the individual empirical Bayesian estimates of clearance based on dose and covariates

Parameter estimates and covariates influence on baseline PVR

Parameter	Unit	Estimate	CV(%)	Transformed value
Fixed Effects				
BASE	1	6.97	1	1064 dyne.s.cm-5
$BASE_{CTD}$	1	6.71	1	821 dyne.s.cm-5
$BASE_{FC}$	1	0.0444	14	
$BASE_{BSA}$	$1/m^2$	-0.0627	18	
$BASE_{age}$	1/y	-0.00122	22	
E_0	1	0.0607	58	6.07%
$Slope_1$	mL/ng	-0.00506	19	
$Slope_{1,dev\ able}$	mL/ng	-0.00829	24	
Thrs	ng/mL	48.9	0	
$Slope_2$	mL/ng	-0.000525	42	
Inter-individual variability (std deviation)				
Adults / Pediatrics	1	0.47 / 0.57	5 / 6	47% / 57%
Residual variability (std deviation)				
Adults / Pediatrics	1	0.22 / 0.336	10 / 5	22% / 33.6%

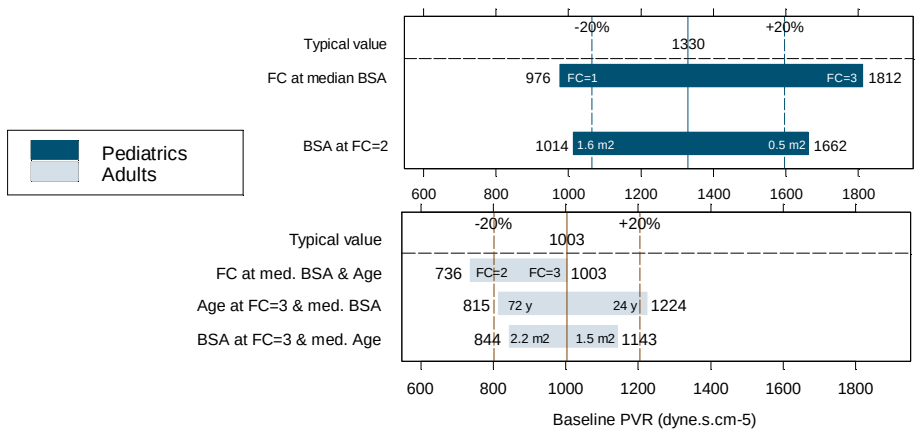
Pediatrics
Adults
Typical value
FC at median BSA
BSA at FC=2
-20%
+20%
1330
976
FC=1
FC=3
1812
1014
1.6 m2
0.5 m2
1662
Typical value
FC at med. BSA & Age
Age at FC=3 & med. BSA
BSA at FC=3 & med. Age
1003
736
FC=2
FC=3
815
72 y
24 y
1224
844
2.2 m2
1.5 m2
1143
600
800
1000
1200
1400
1600
1800
Baseline PVR (dyne.s.cm-5)

RESULTS

Exposure-effect relationship in adults and in pediatric patients

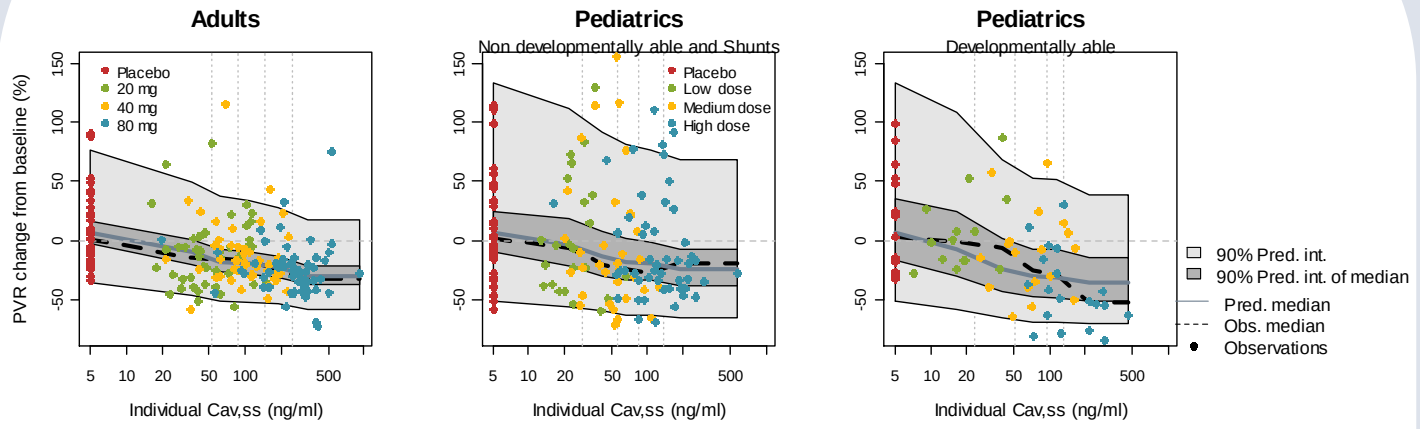
Adults
Pediatrics
Non developmentally able and Shunts
Pediatrics
Developmentally able
Placebo
20 mg
40 mg
80 mg
Low dose
Medium dose
High dose
PVR change from baseline (%)
Individual Cav,ss (ng/ml)
90% Pred. int.
90% Pred. int. of median
Pred. median
Obs. median
Observations

Distribution of PVR improvement measured as change from baseline

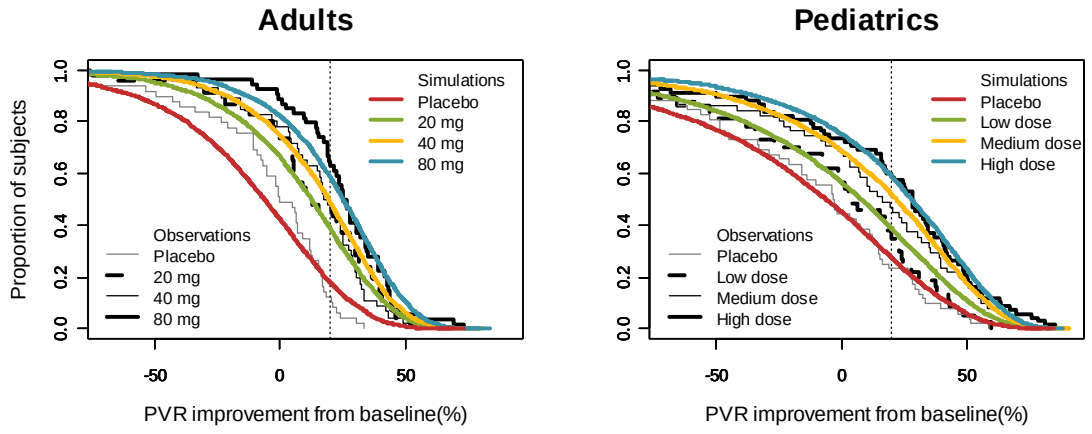
Adults
Pediatrics
Proportion of subjects
Simulations
Placebo
20 mg
40 mg
80 mg
Low dose
Medium dose
High dose
Observations
PVR improvement from baseline(%)

Simulations in pediatrics

Model based simulations suggested that for children a dose of 10 mg TID up to 20 kg and 20 mg TID beyond achieves a comparable PVR response to adults at the labeled dose of 20 mg TID i.e. a 20% improvement in change from baseline in 40% of patients. Clinical results and similar analysis [8, 9] on pVO2 data in children 7-17 y confirmed the selected regimen.

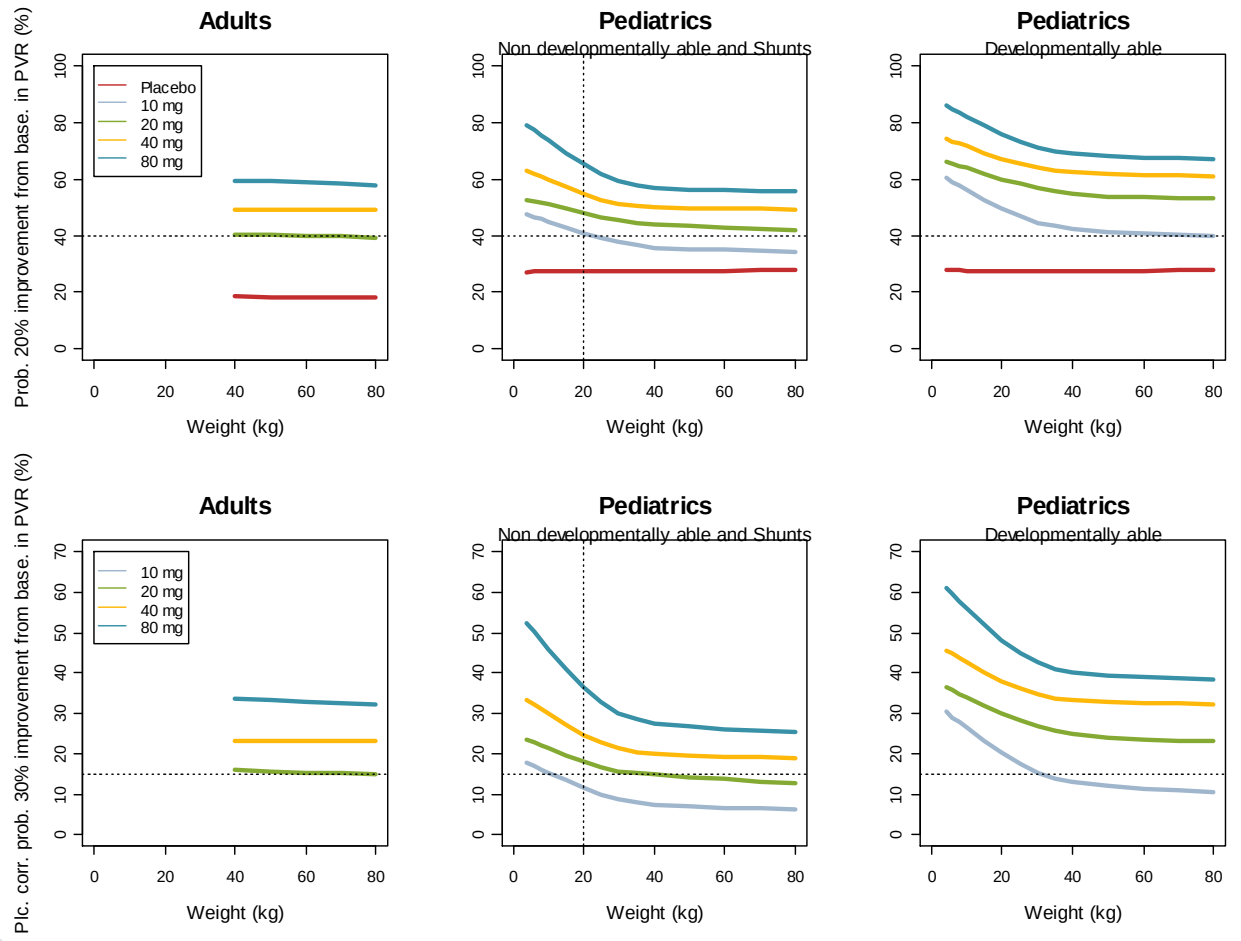
Adults
Pediatrics
Non developmentally able and Shunts
Pediatrics
Developmentally able
Prob. 20% improvement from base. in PVR (%)
Plc. corr. prob. 30% improvement from base. in PVR (%)
Placebo
10 mg
20 mg
40 mg
80 mg
Weight (kg)

CONCLUSIONS

A recent FDA assessment of the relationship between 6MWD and PVRI proposes the use of hemodynamic endpoints to support drug development in PAH especially in the pediatric population. Prior to this analysis, the FDA assessment was leveraged and contrasted with Pfizer's sildenafil data.

A model developed for PVR described the baseline data, the exposure-response relationship and the entire distribution of PVR improvements in both population: adults and pediatrics.

Utilizing model based simulations of PVR outcomes allowed bridging efficacy from adults to children supporting dose recommendations for sildenafil in pediatric PAH patients of 10 mg TID below 20 kg body weight and 20 mg TID beyond.

REFERENCES

[1] L Harnisch et al. Hemodynamics as a substitute for exercise capacity endpoints in patients with pulmonary arterial hypertension (PAH): A model based assessment for sildenafil, ERS, Barcelona, 2010, Presentation 200.

[2] J Wagg, et al. Exploratory modeling of exercise capacity and hemodynamic endpoints in patients with pulmonary arterial hypertension, ESC, Stockholm, 2010, Presentation 86389.

[3] FDA Cardiovascular and Renal Drugs Advisory Committee Meeting, July 29, 2010. http://www.fda.gov/AdvisoryCommittees/Calendar/ucm217266.htm

[4] L Harnisch et al. Hemodynamics as an additional measure to prove efficacy and to provide a dose rationale for sildenafil in pediatric patients with pulmonary arterial hypertension (PAH) , ATS, Denver Int. Conference, 2011.

[5] Galie et al, Sildenafil Citrate Therapy for Pulmonary Arterial Hypertension, N Engl J Med, 2005;353:2148-57.

[6] Simonneau et al, Addition of Sildenafil to Long-Term Intravenous Epoprostenol Therapy in Patients with Pulmonary Arterial Hypertension, Ann Intern Med, 2008;149:521-530.

[7] S Watt et al.Population pharmacokinetics of sildenafil in paediatric and adult patients with pulmonary arterial hypertension, ESC, Stockholm, 2010, Poster 83747.

[8] N Hayashi et al. Population PK of Sildenafil and PK/PD assessment of Exercise tolerability in children with Pulmonary Arterial Hypertension (PAH), PAGE, St Petersburg, 2009, Abstract 1523.

[9] L Harnisch et al. Exercise tolerability in children with pulmonary arterial hypertension (PAH), a population PK/PD assessment of the effects of sildenafil, ERS, Vienna, 2009, Poster 3890.